2024-A-12247- LB-ENDO

Assessing the Effectiveness of GluCare's Hybrid Model of Care in Diabetes and Chronic Kidney Disease Management

Hadoun Jabri [1], **Yousef Said** [1], **Hala Zakaria** [1], **Sheikha Alshehhi** [1], **Mohamad Alhosaini** [2], **Noah Almarzooqi** [1], **Ali Hashemi** [1], **Ihsan Almarzooqi** [1]

1. GluCare.Health, United Arab Emirates, Dubai, Al Wasl Road, Jumeirah 1, hadoun@glucare.health, yousef@glucare.health, hala@glucare.health, sheikha@glucare.health, ali@glucare.health, ihsan@glucare.health
2. Sheikh Khalifa General Hospital, United Arab Emirates, Umm Al Quwain, Mnalhosaini@gmail.com

1 Background

Diabetes remains one of the leading causes of chronic kidney disease among other complications worldwide, necessitating the need to explore new approaches to monitor and manage them. Traditional, in-clinic care models are largely episodic, whilst digital-only diabetes solutions have recently been shown to be of limited utility (1). Both models lack the continuous monitoring of an interdisciplinary team required for effective management of chronic diseases.

2 Objectives

GluCare Health in Dubai, UAE, practices a hybrid care model that is a vertically integrated model and includes the management of both in-clinic care with continuous remote digital monitoring and engagement.The aim of this study is to evaluate its effectiveness in improving outcomes for diabetic patients with CKD.

3 Methodology

A retrospective study of 51 T2D patients receiving anti-diabetic medications and actively managed via the GluCare.Health hybrid model for 6 months. This included an in-person visit at baseline and every 3 months alongside continuous digital monitoring and guidance on medication compliance, diet modification, and life coaching, complemented by bi-weekly contacts from the clinical team—including physicians, dietitians, and coaches for consistent support and guidance. Laboratory tests were performed at each follow-up visit.

Overview of the GluCare Hybrid Journey

The GluCare.health Hybrid Journey represents an innovative approach in diabetes management by integrating digital health tools with traditional in-person care. This model utilizes wearable devices to continuously monitor patients' vital signs and glucose levels, enabling real-time data analysis.

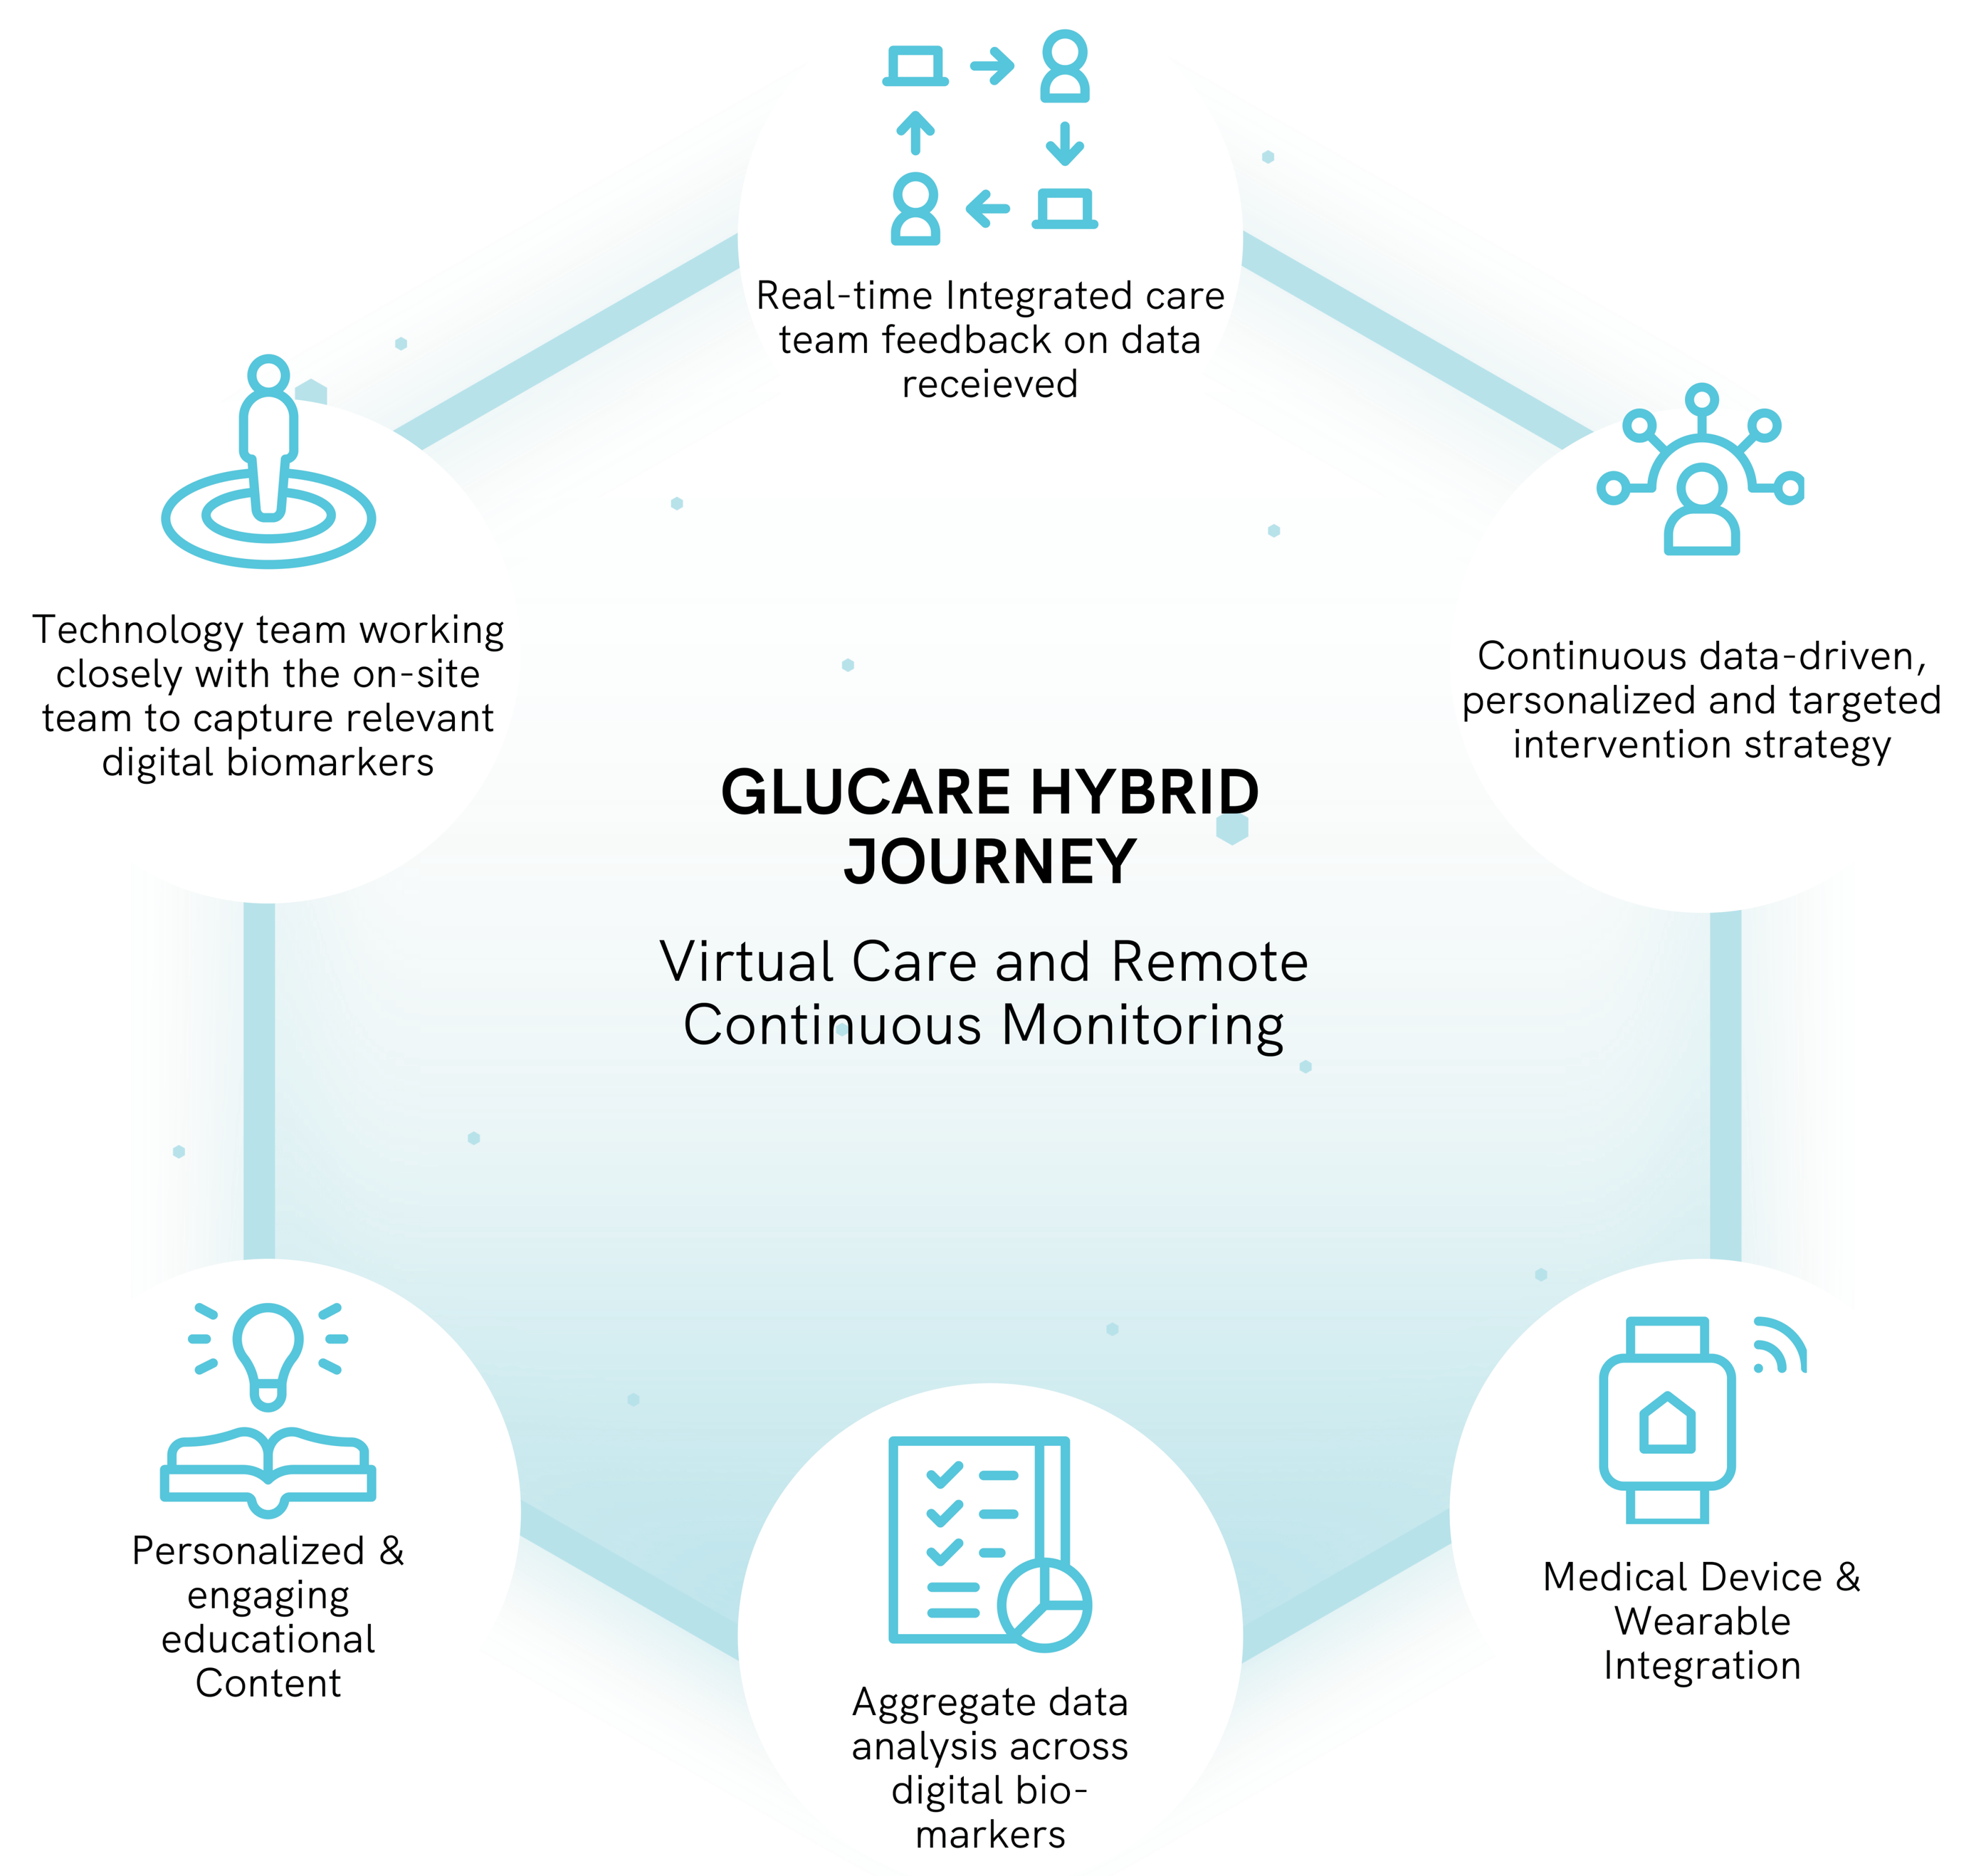
Real-time Integrated care team feedback on data receieved
Technology team working closely with the on-site team to capture relevant digital biomarkers
Continuous data-driven, personalized and targeted intervention strategy
GLUCARE HYBRID JOURNEY
Virtual Care and Remote Continuous Monitoring
Personalized & engaging educational Content
Aggregate data analysis across digital bio-markers
Medical Device & Wearable Integration

Figure 1: Overview of the GluCare.Health Hybrid Model of Care

4 Results

Decrease in weight	Decrease in Waist Circumference	Reduction in Cardiovascular Risk
7% ($p<0.001$)	7% ($p<0.05$)	43% ($p<0.05$)

Improvements in GlyCemic Control and ACR

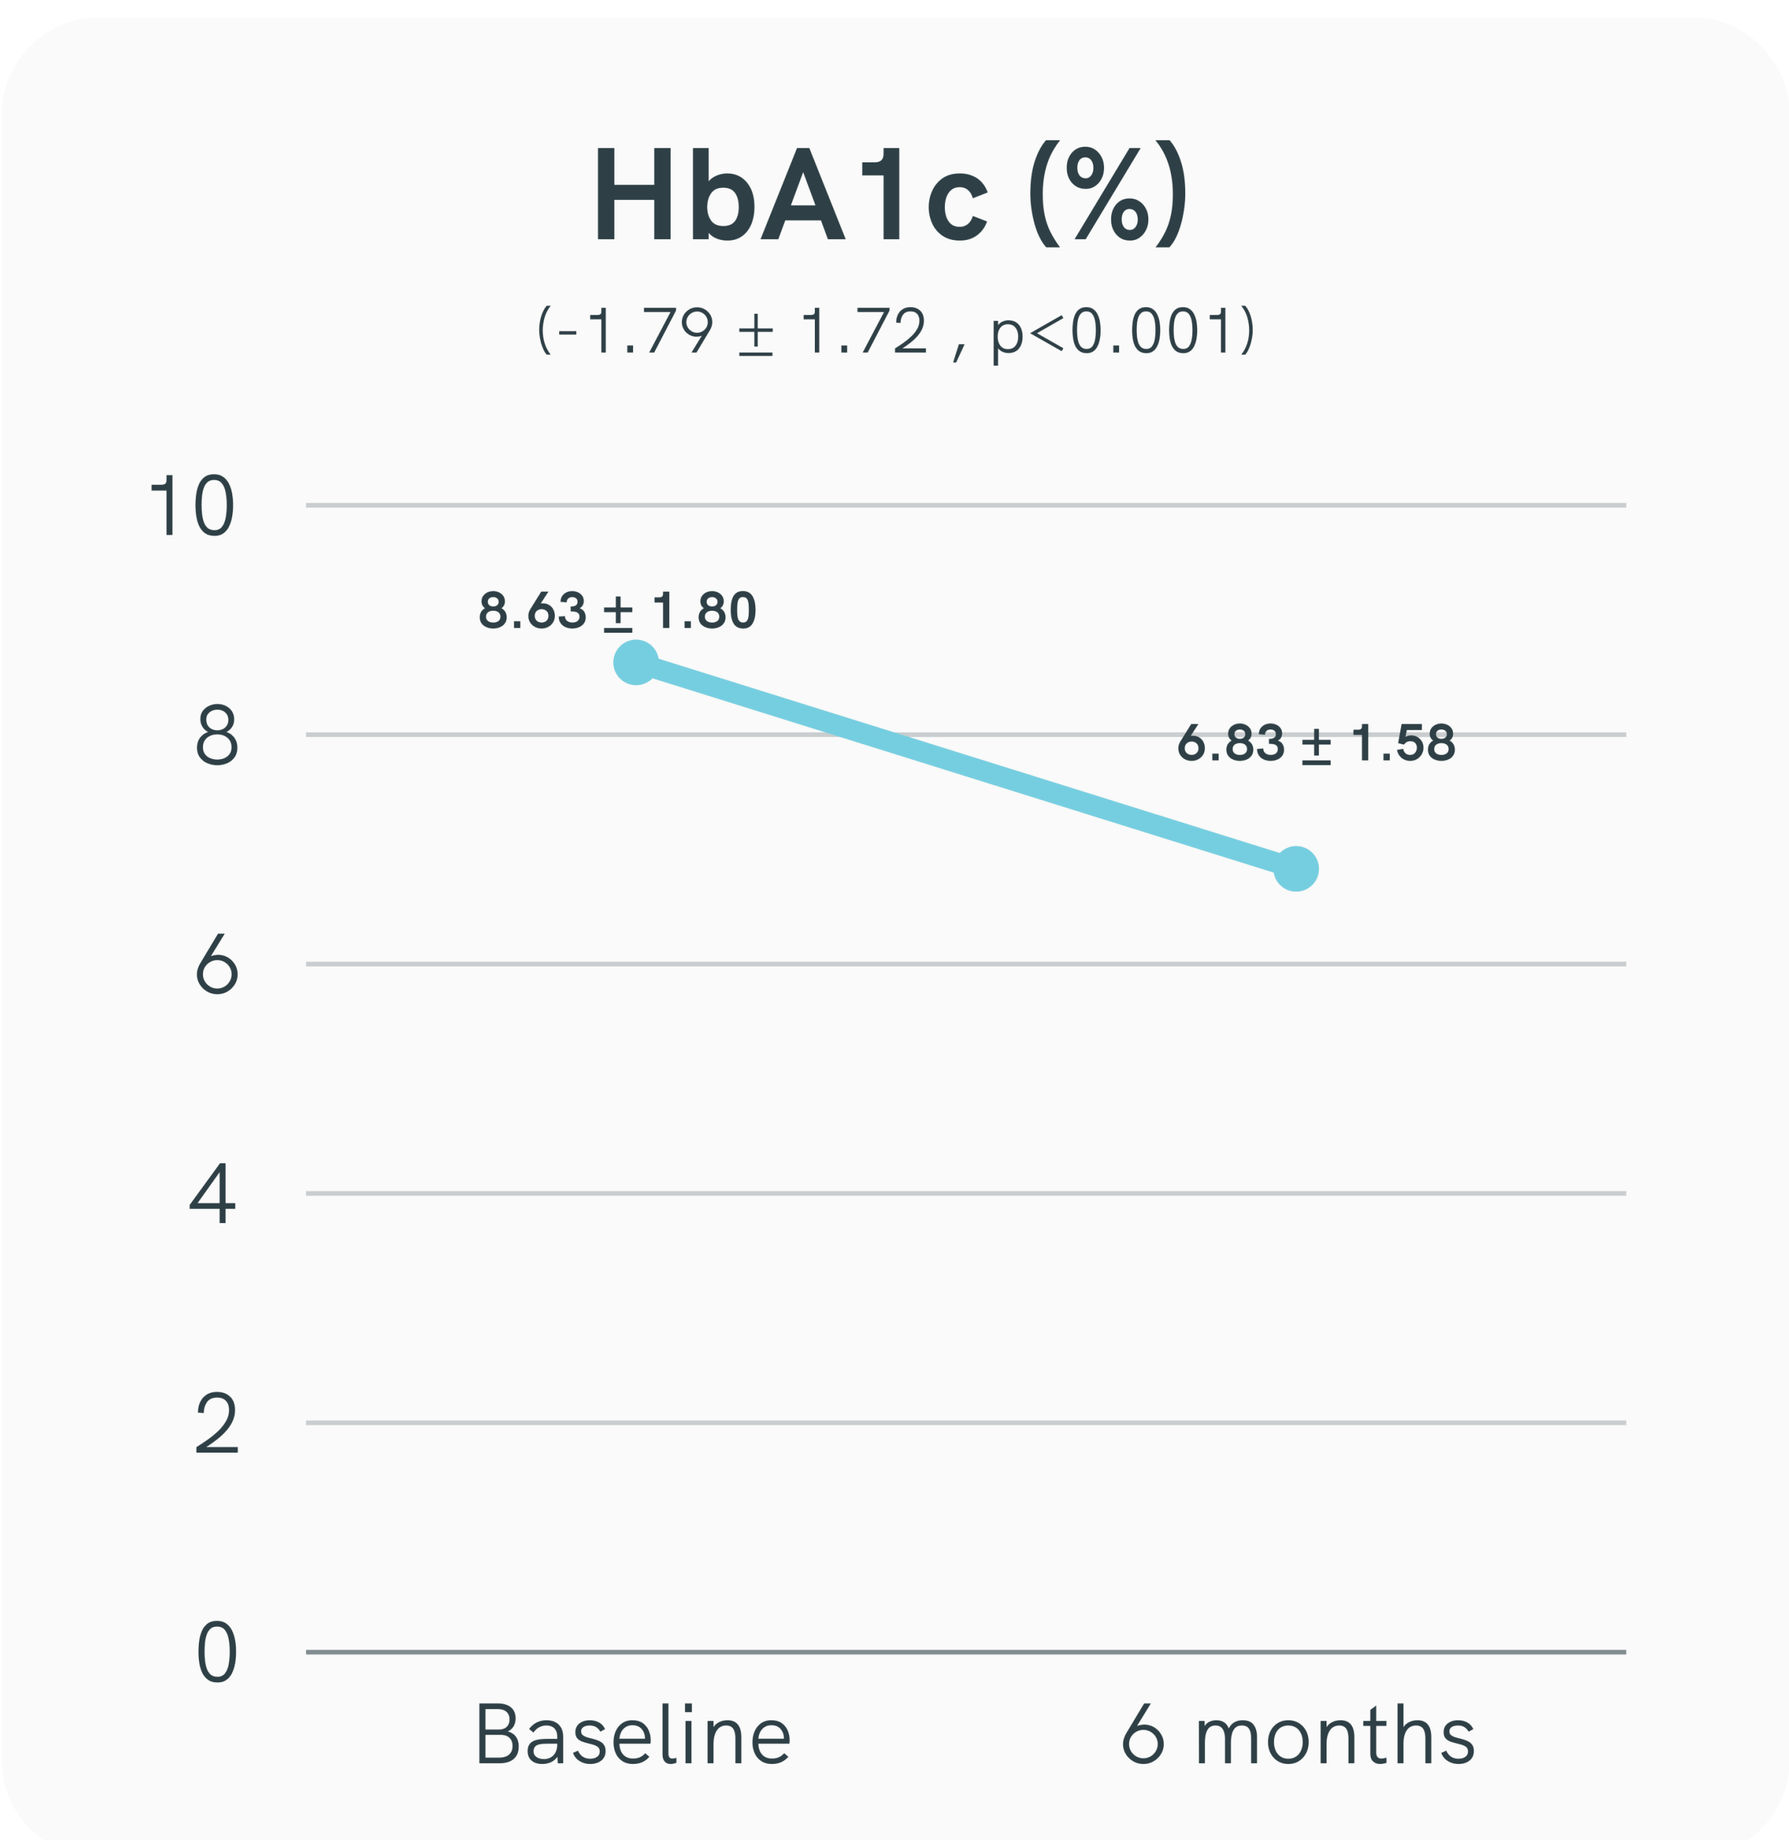
HbA1c (%)
(-1.79 ± 1.72 , p<0.001)
10
8
6
4
2
0
8.63 ± 1.80
6.83 ± 1.58
Baseline
6 months

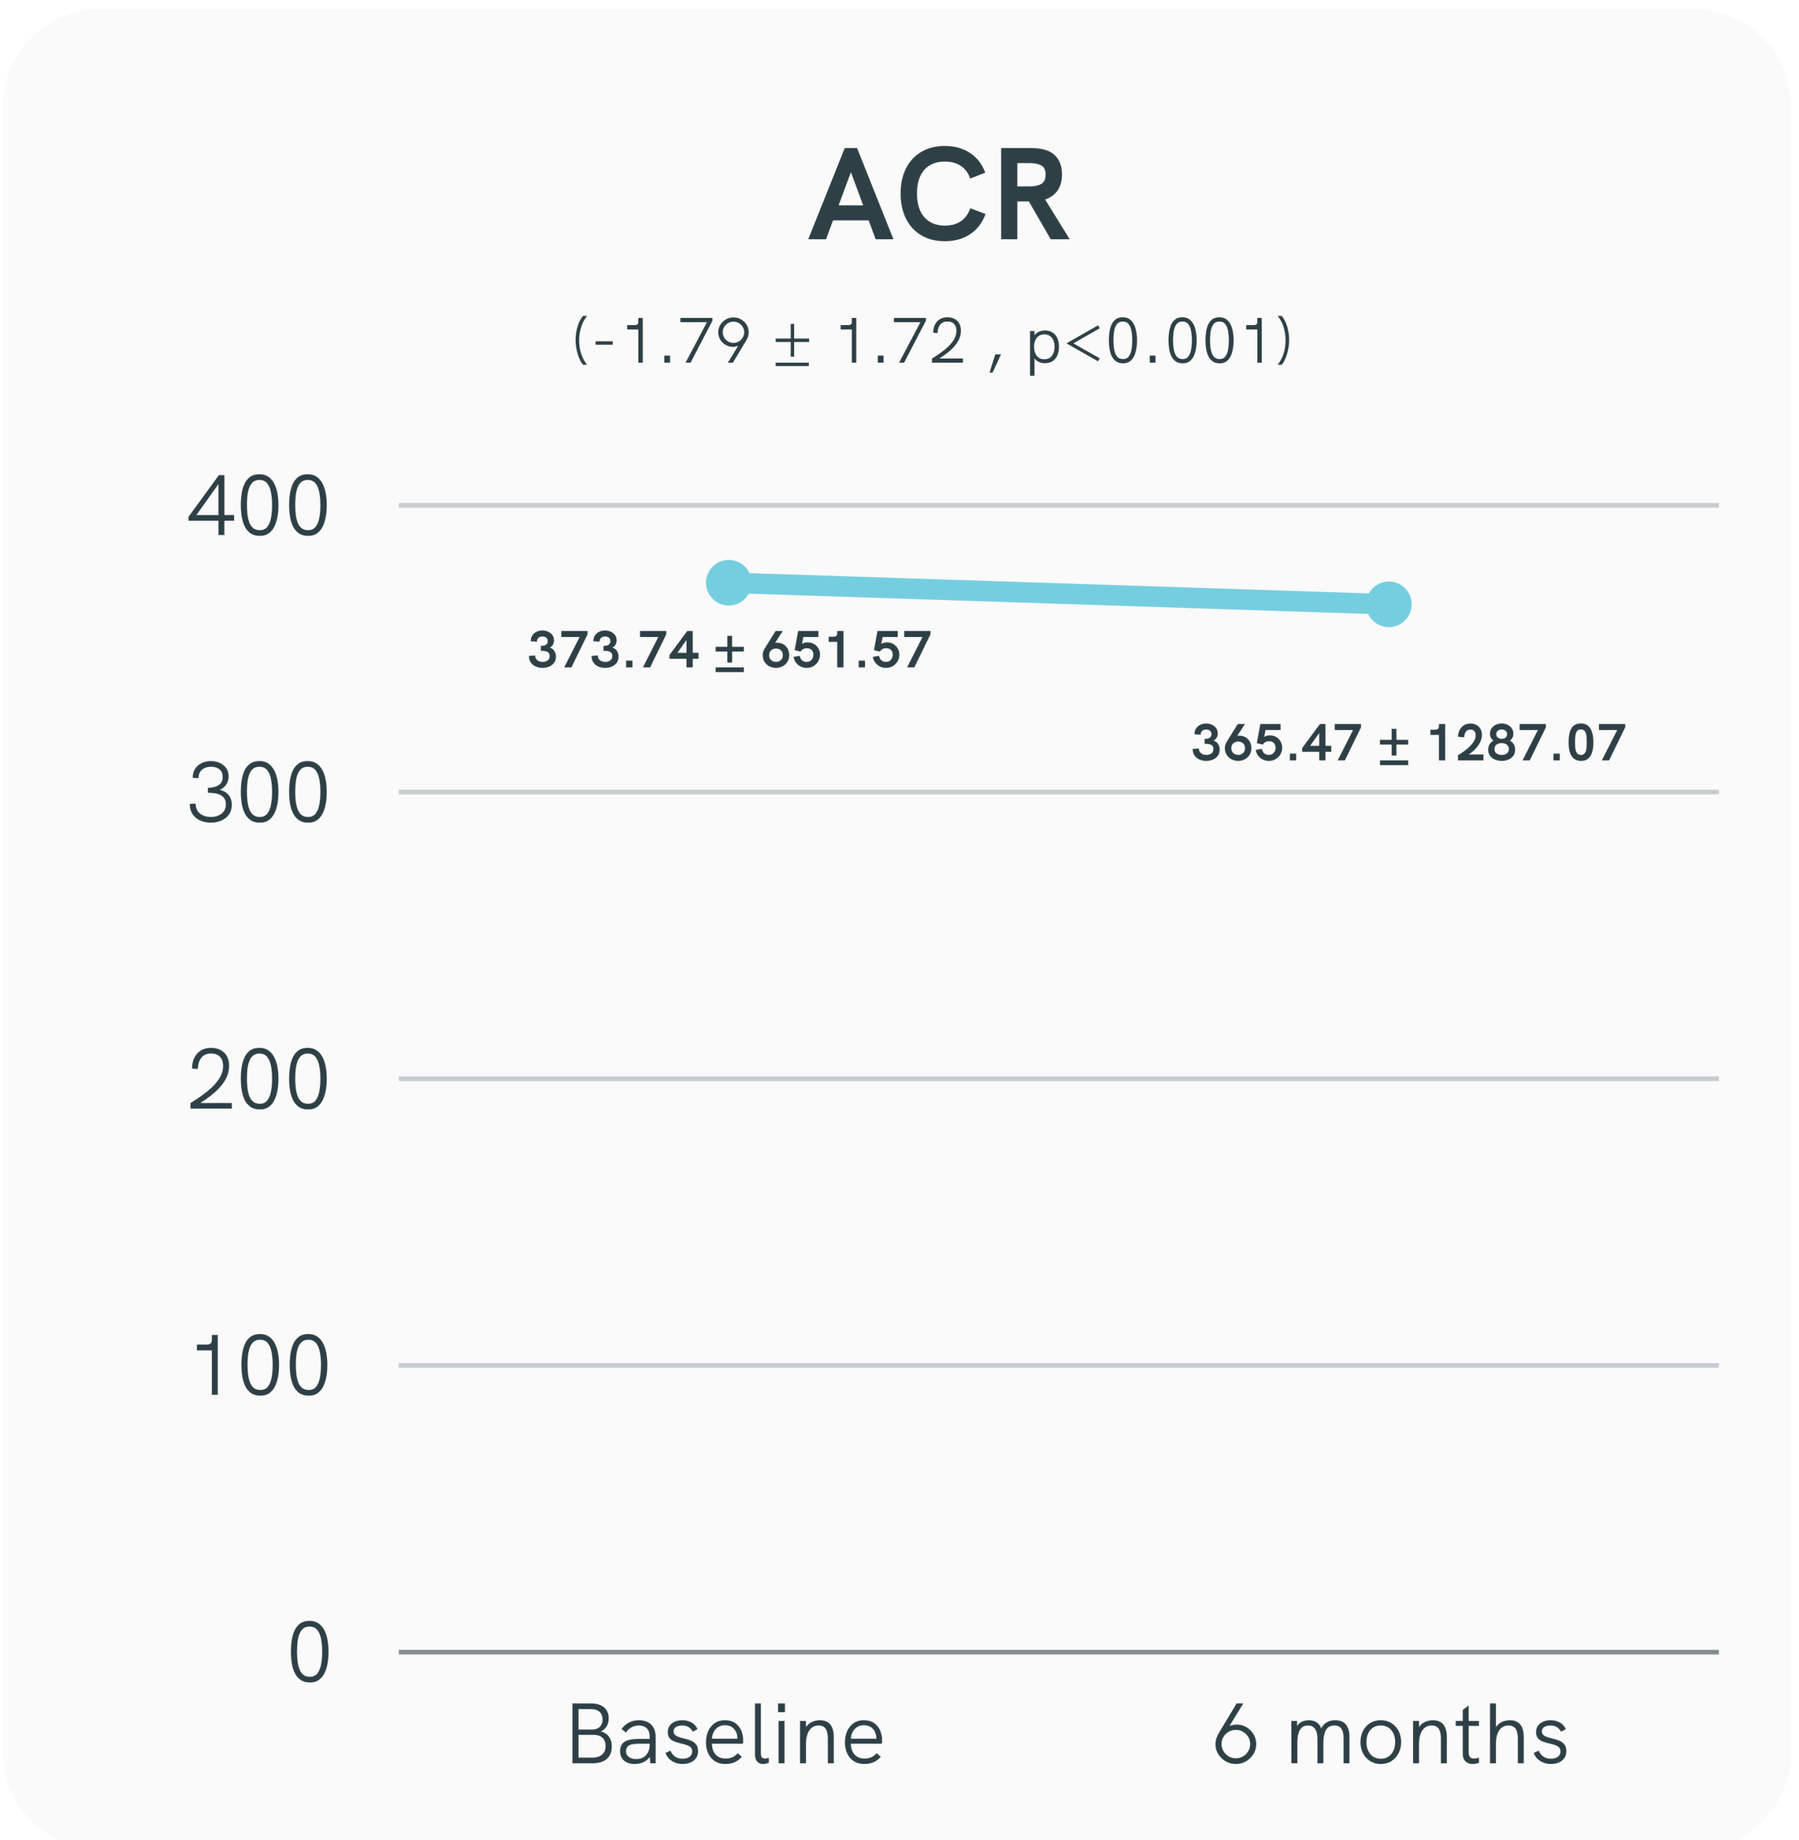
ACR
(-1.79 ± 1.72 , p<0.001)
400
300
200
100
0
373.74 ± 651.57
365.47 ± 1287.07
Baseline
6 months

Improvements in Blood Pressure

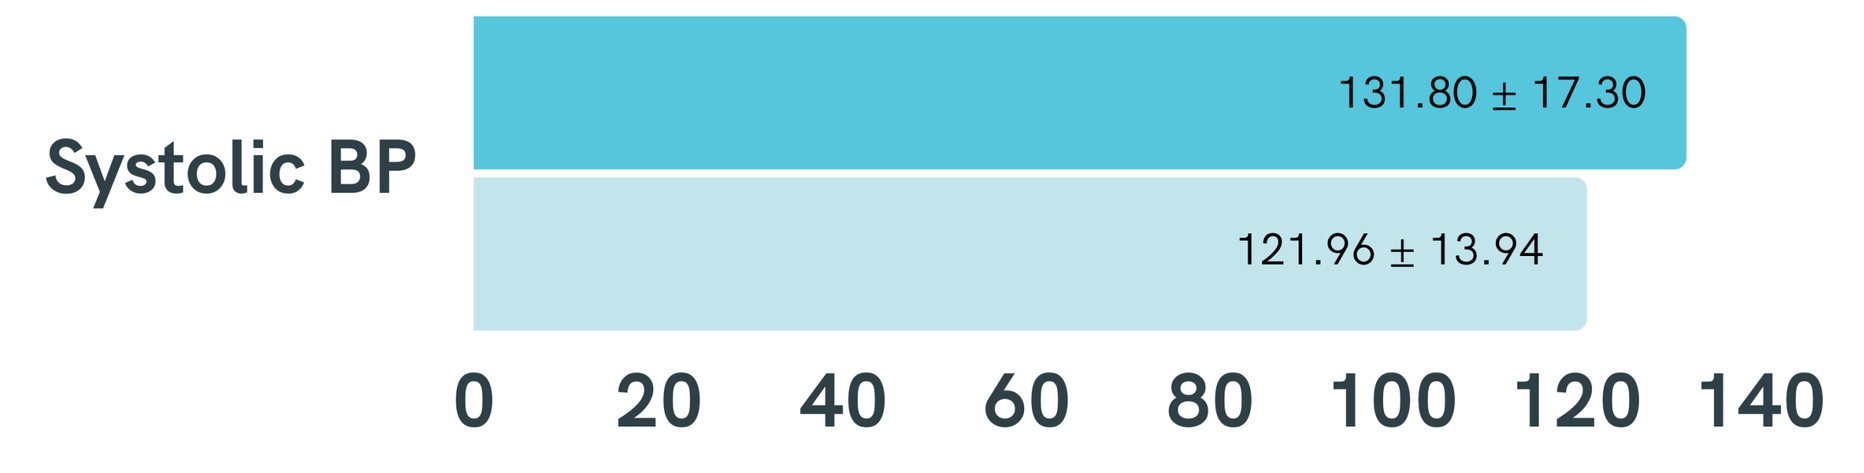
Systolic BP
131.80 ± 17.30
121.96 ± 13.94
0 20 40 60 80 100 120 140

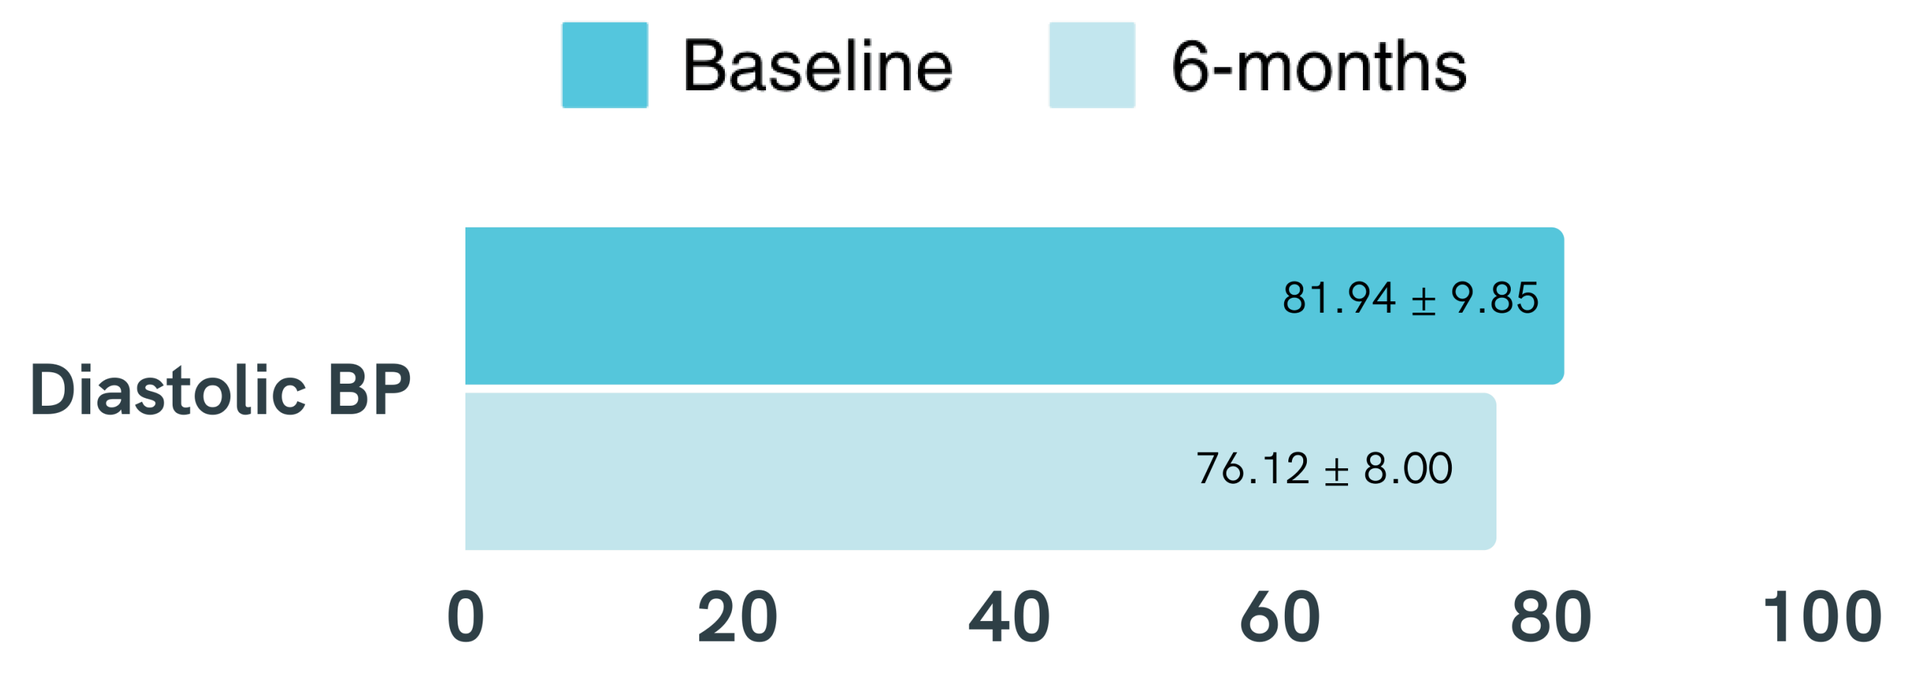
Baseline
6-months
Diastolic BP
81.94 ± 9.85
76.12 ± 8.00
0 20 40 60 80 100

5 Interpretation of the results and conclusions

- These outcomes highlight the potential of new care models that combine in-clinic care with continuous monitoring and engagement by a multidisciplinary team to enhance disease management.
- Notable blood pressure reductions were achieved without increasing medication. A 10 mmHg drop in systolic BP is more than the expected drop due to weight loss alone, suggesting the overall intervention had a cumulative effect. Despite minimal ACR improvements, the outcomes are promising given the management's brief duration.
- Further studies are needed to assess long-term renal effects.The findings support the use of hybrid care in chronic disease management and demonstrates the ability of hybrid care to improve critical health metrics.

References: 1. Digital Diabetes Management Solutions. (n.d.). Retrieved April 4, 2024, from https://phti.com/assessment/digital-diabetes-management-tools/